DEVELOPMENT OF A SUCCESSFUL LYOPHILIZATION PROCESS FOR LENTIVIRAL VECTOR CLINICAL BATCHES

THERAVECTYS, 1 mail du Professeur Georges Mathé,
94800 VILLEJUIF, FRANCE

Nicolas DELACROIX, Alice PAILLERET, Guillaume DEPLAINE, Julien DEROYER, Cécile BAUCHE

BACKGROUND

THERAVECTYS is a privately-owned, fully integrated discovery and clinical development biotech company originating from the **Pasteur Institute**. The company capitalizes on over 15 years of fundamental research in the field of **lentiviral vectors** and owns over **20 patent families**, either developed in-house or exclusively licensed from worldwide renown institutions from which the company has secured **exclusive worldwide rights for the use of lentiviral vectors in vaccination and immunotherapy applications**.

Based on its lentiviral vector technology platform, THERAVECTYS develops **therapeutic vaccines** and **immunotherapies** to fight cancers and infectious diseases, including a proprietary and **differentiated CAR T-cell technology platform**.

The company has completed the first-ever vaccination clinical trial conducted with lentiviral vectors confirming both the **safety & immunogenicity** of the lentiviral vector platform.

THERAVECTYS has recently been granted the status of a **GMP pharmaceutical manufacturing establishment**, by the French regulatory agency to produce lentiviral vectors for clinical development purposes and to manipulate human cells in the context of **Chimeric Antigen Receptors (CARs) and T-Cell Receptor (TCR)-based cell therapies**.

THERAVECTYS aims at ensuring the stability of its lentiviral vector batches to be compatible with the constraints of a global supply chain.

One approach to secure the stability of viral vector based products is **lyophilization** (or freeze-drying).

THERAVECTYS has developed a unique method for the **freeze-drying** of its clinical grade batches which allows the storage of the final product at -20°C or ideally at +4°C while maintaining the characteristics of the vector particles after reconstitution and their compatibility with a **direct injection into humans** or any other clinical applications (T-cell modification, gene therapy...).

FORMULATION & LYOPHILIZATION TRIALS

- Stable formulation
- Preserved immunogenicity
- Controled impurities profile

Lyophilized lentivector compatible with administration to human.

A **formulation screening** of different lyo-protectants show the THERAVECTYS lyophilisation process relevance for the successful storage of lentiviral vectors preparations with the viral titer yield conservation. Comparing data obtained at +4°C and -20°C storage conditions *(fig.1, fig.2)*, the presence of a concentrated disaccharide matrix show the reliability of the freeze-drying process to stabilize preparations as shown with D1 and D7 results after manufacturing. -20°C data s+4°C data shows the well-known residual humidity impact when water is in liquid form along the 45 days storage compared to -20°C data when residual water is in its solid form.

COMPARISON OF DIFFERENT LYOPROTECTANTS

Fig. 1

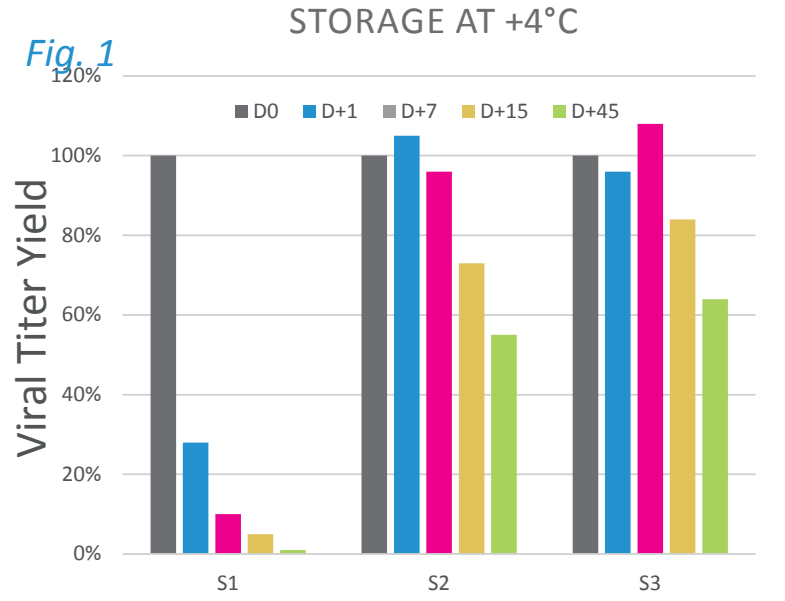
STORAGE AT +4°C
D0
D+1
D+7
D+15
D+45
Viral Titer Yield
120%
100%
80%
60%
40%
20%
0%
S1
S2
S3

Fig. 2

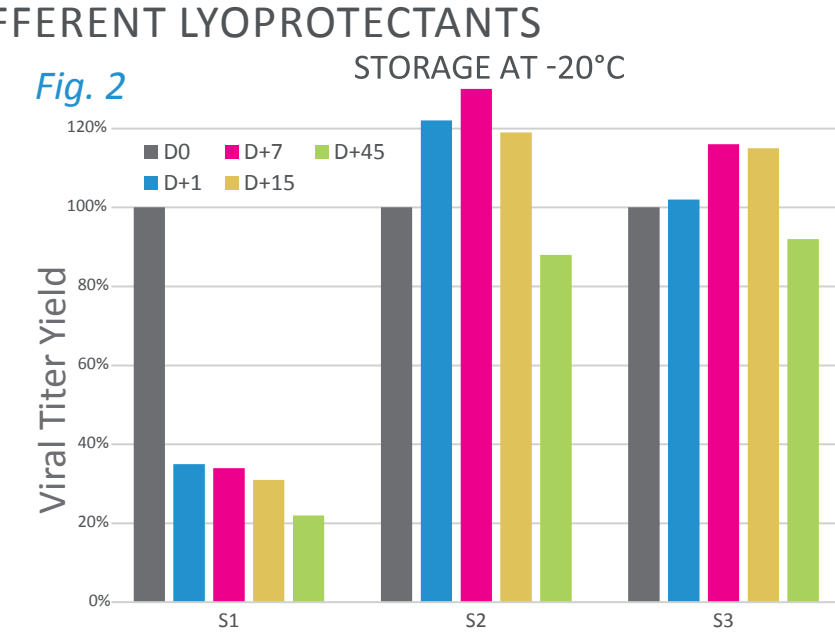
STORAGE AT -20°C
D0
D+1
D+7
D+15
D+45
Viral Titer Yield
120%
100%
80%
60%
40%
20%
0%
S1
S2
S3

S1: No lyoprotectant - S2: Sucrose 170 g/L - S3: Trehalose 170 g/L

Furthermore, to allow a better colloidal stabilization of lentiviral preparation along purification process, Human serum albumin is added to solutions and a stability study encompass. Results *(fig. 3)* confirms the relevance of the THERAVECTYS original lyophilisation process to store lentivirus with potency conservation.

Fig. 3

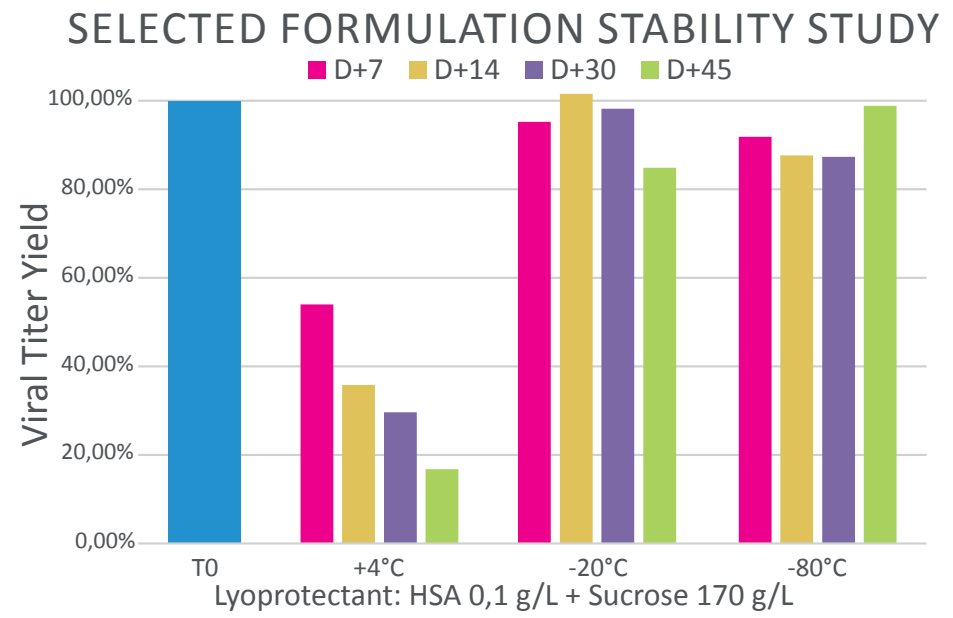
SELECTED FORMULATION STABILITY STUDY
D+7
D+14
D+30
D+45
Viral Titer Yield
100,00%
80,00%
60,00%
40,00%
20,00%
0,00%
T0
+4°C
-20°C
-80°C

Lyoprotectant: HSA 0,1 g/L + Sucrose 170 g/L

Lyophilisation impact on lentiviral vector profile was evaluated before and after the lyophilization The following parameters was analyzed :

	Before lyophilization	After lyophiliza-tion	At each stability time point
Viral titer	✓	✓	✓
Impurities profile	✓	✓	✓
Immunogenicity	✓	✓	End point

Immunogenicity trials *(fig. 4)* show that freeze-drying had no impact on the breadthand intensity of the cellular immune response.

Fig. 4

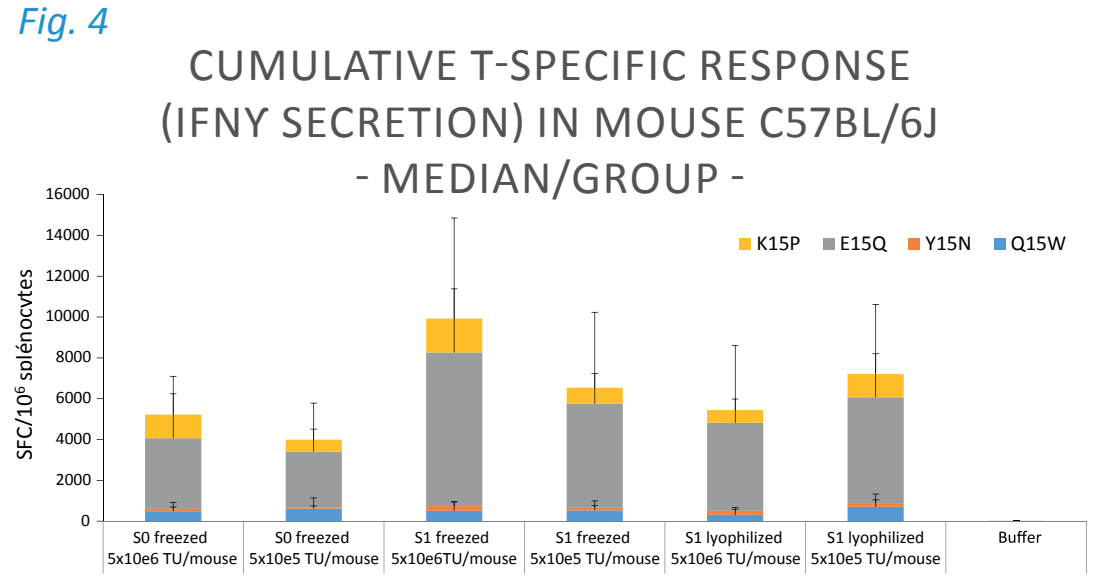
CUMULATIVE T-SPECIFIC RESPONSE
(IFNY SECRETION) IN MOUSE C57BL/6J
- MEDIAN/GROUP -
K15P
E15Q
Y15N
Q15W
SFC/10⁶ splénocytes
16000
14000
12000
10000
8000
6000
4000
2000
0
S0 freezed 5x10e6 TU/mouse
S0 freezed 5x10e5 TU/mouse
S1 freezed 5x10e6TU/mouse
S1 freezed 5x10e5 TU/mouse
S1 lyophilized 5x10e6 TU/mouse
S1 lyophilized 5x10e5 TU/mouse
Buffer

S0: No lyoprotectant S1: Sucrose 170g/l + BSA

QC testing was done on these freeze-driedproducts and showed that the lyophilisationprocess did not alter the content in terms ofimpurities. The products still meet the regulatory agenciescompliance for clinical trials even after thelyophilisation process.

Even though the evaluation on long-term stability is still on-going, all formulations tested show that the product can be stored and distributed at -20°C (6 months stability results available beginning of August). Simply being able to store the product at -20°C instead of the recommended -80°C opens up a large array of possibilities in terms of distribution and is therefore a definite added value.

PROCESS DESIGN

THERAVECTYS has designed the following process in order to obtain a GMP clinical-grade final product in a defined formulation that allows the lyophilization of lentiviral vectors either produced in adherent cells cultures (with FBS) or in suspension cells cultures (grown in synthetic culture medium without FBS).

The vectors are protected during freeze-drying by the addition of a lyoprotectant during the purification process and stability is evaluated over time.

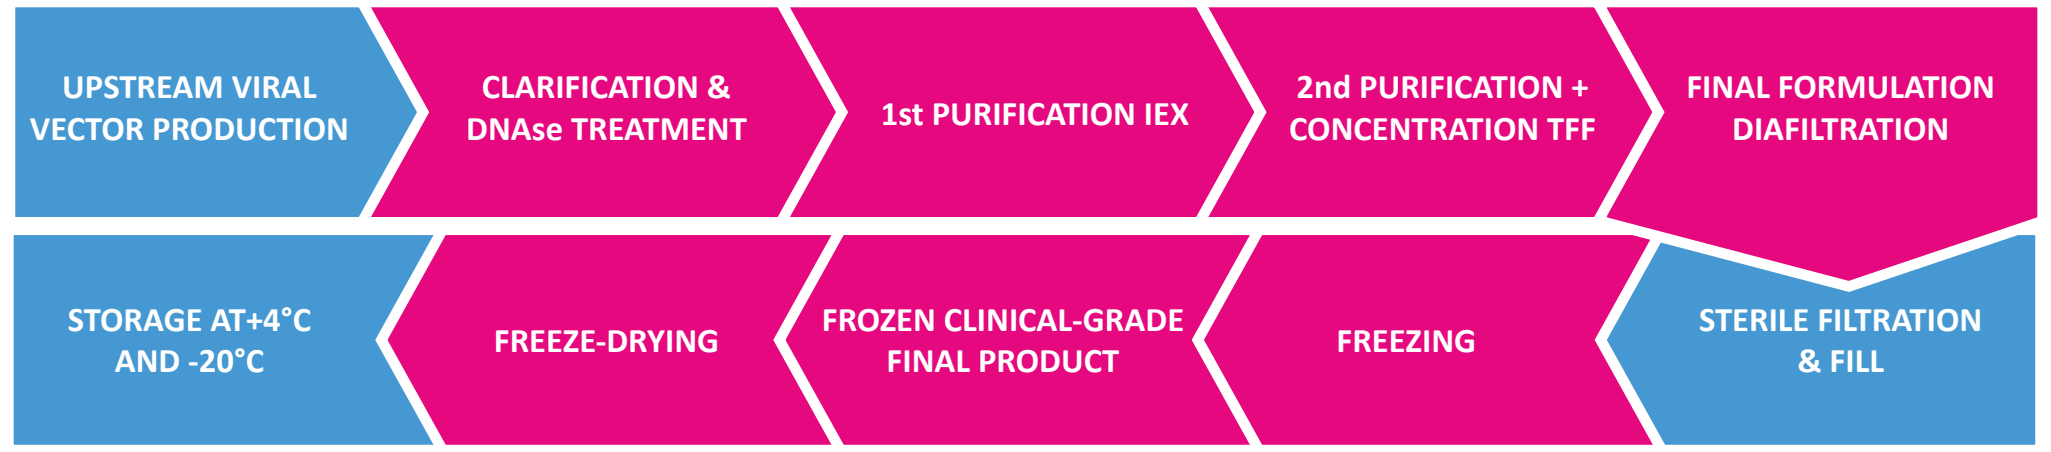
UPSTREAM VIRAL VECTOR PRODUCTION
CLARIFICATION & DNAse TREATMENT
1st PURIFICATION IEX
2nd PURIFICATION + CONCENTRATION TFF
FINAL FORMULATION DIAFILTRATION
STERILE FILTRATION & FILL
FREEZING
FROZEN CLINICAL-GRADE FINAL PRODUCT
FREEZE-DRYING
STORAGE AT+4°C AND -20°C

NEXT STEPS

Not only is THERAVECTYS the first company to ever obtain a freeze-dried, clinical-grade lentiviral vector product, THERAVECTYS has also demonstrated that our freeze-drying process enables a sufficient conservation of our product's biological activity to counter the distribution and storage limitations often met when undertaking large scale clinical trials. Nevertheless, ongoing development still remains in order for us to reach our ultimate goal of being able to store a stable clinical-grade product for up to 3 years at +25°C under a %RH of 60%.

Our current studies are based on finding the best conditions for the freezing step of our lyophilization process but also on finding the optimal formulation prior to freeze-drying (which lyoprotectants to use and at which concentrations) that allows a long-term storage whilst maintaining our product's safety and efficacy.

All our studies are done in view of a future industrialization of our freeze-drying process and so all our choices in terms of freezing, formulation and process lengths are done following the constraints of a GMP large-scale production.

© Copyright 2015 THERAVECTYS. All Rights reserved.